Repulsive bias in category formation: Beyond confirmation bias

Long Ni, Lucas Caceres, & Michael S. Landy

Department of Psychology and Center for Neural Science, New York University, NY 10003

Question

How do we estimate category means?

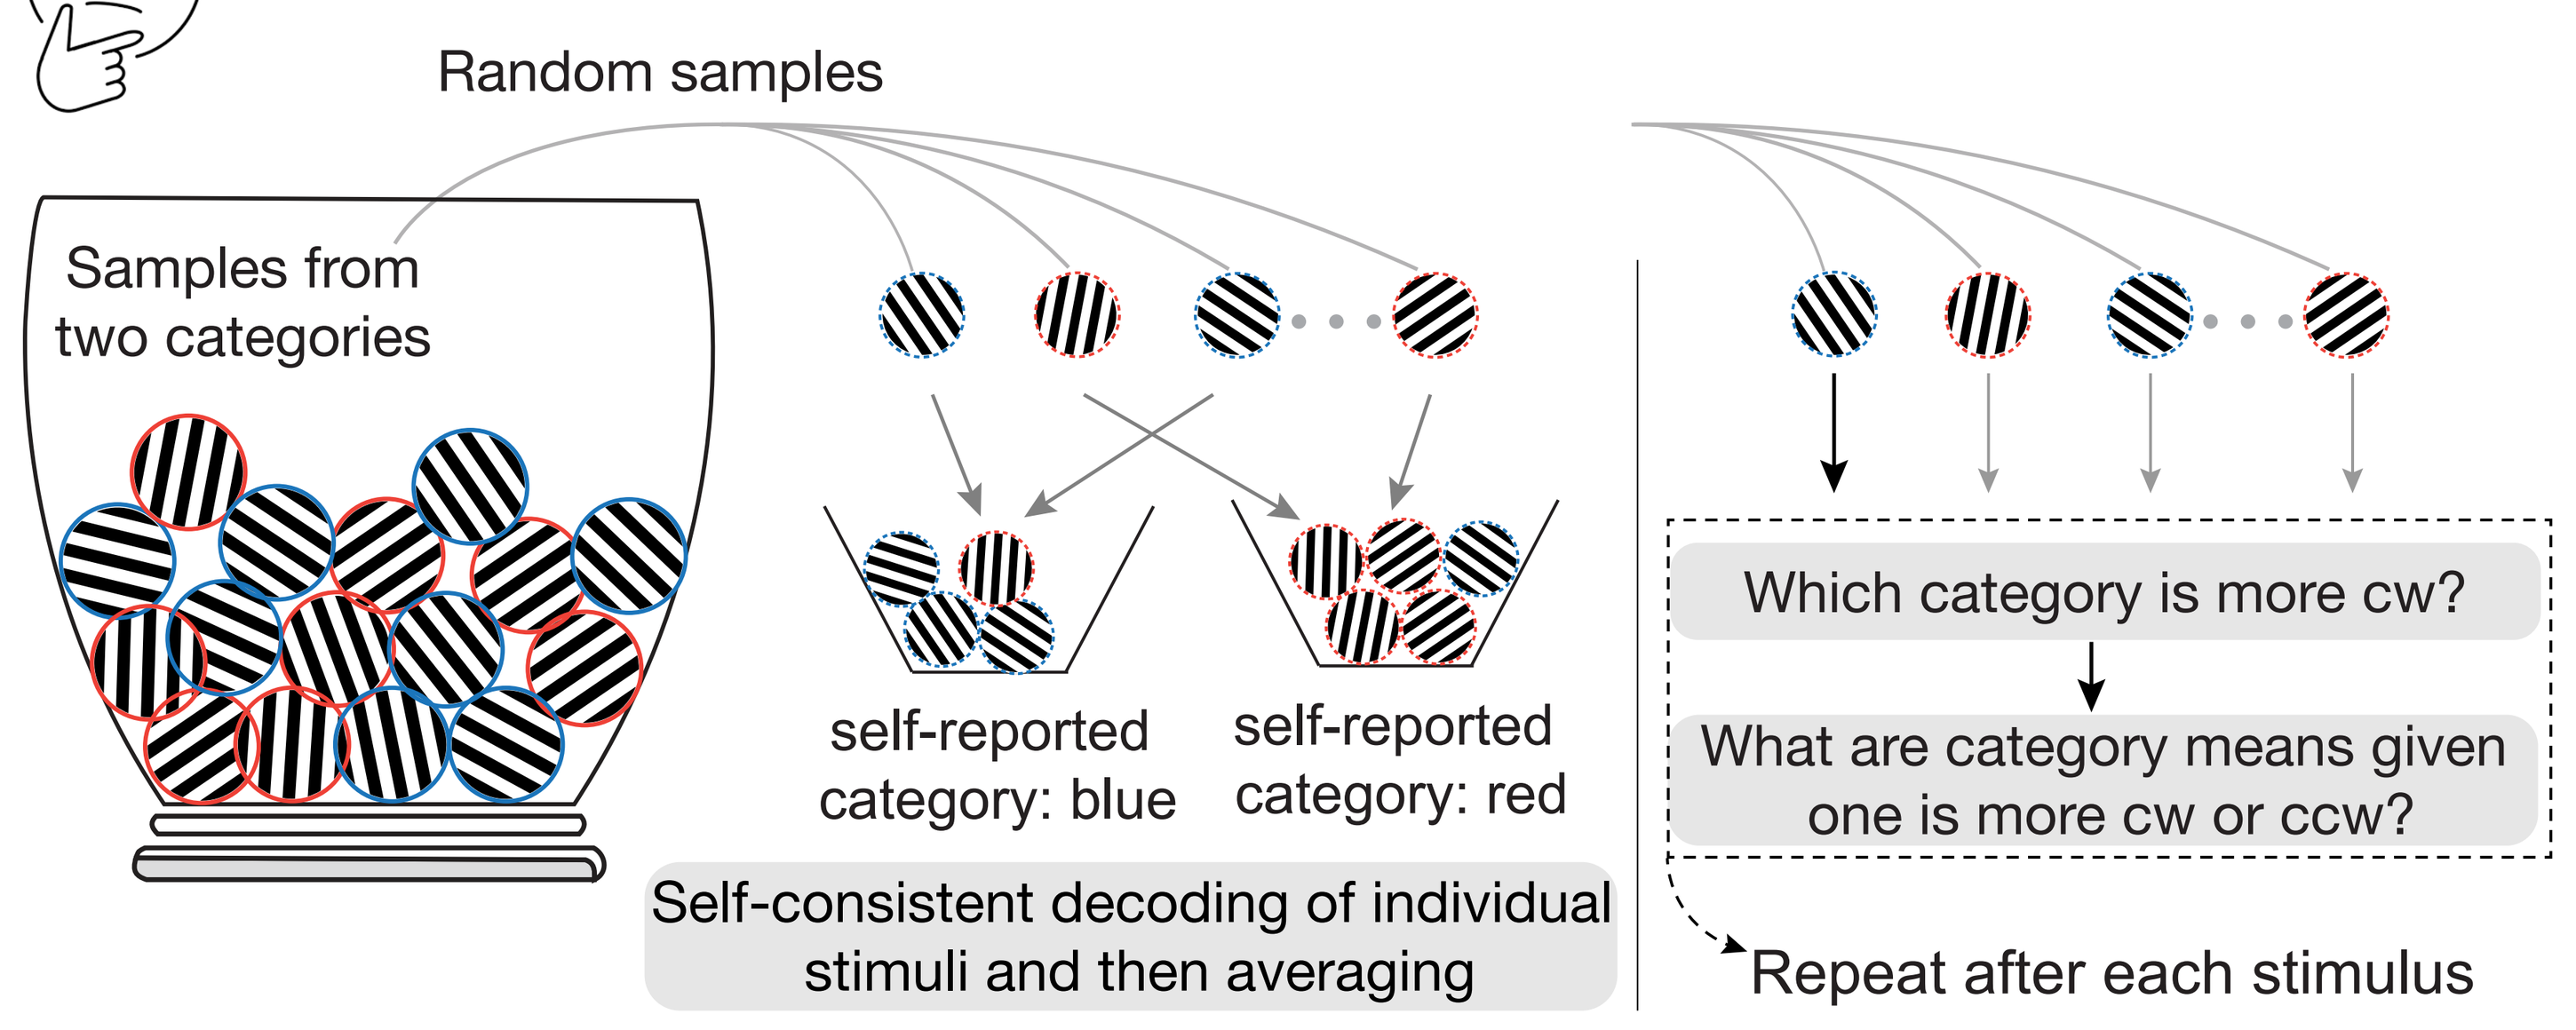
Random samples
Samples from two categories
self-reported category: blue
self-reported category: red
Self-consistent decoding of individual stimuli and then averaging
Which category is more cw?
What are category means given one is more cw or ccw?
Repeat after each stimulus

Experiment

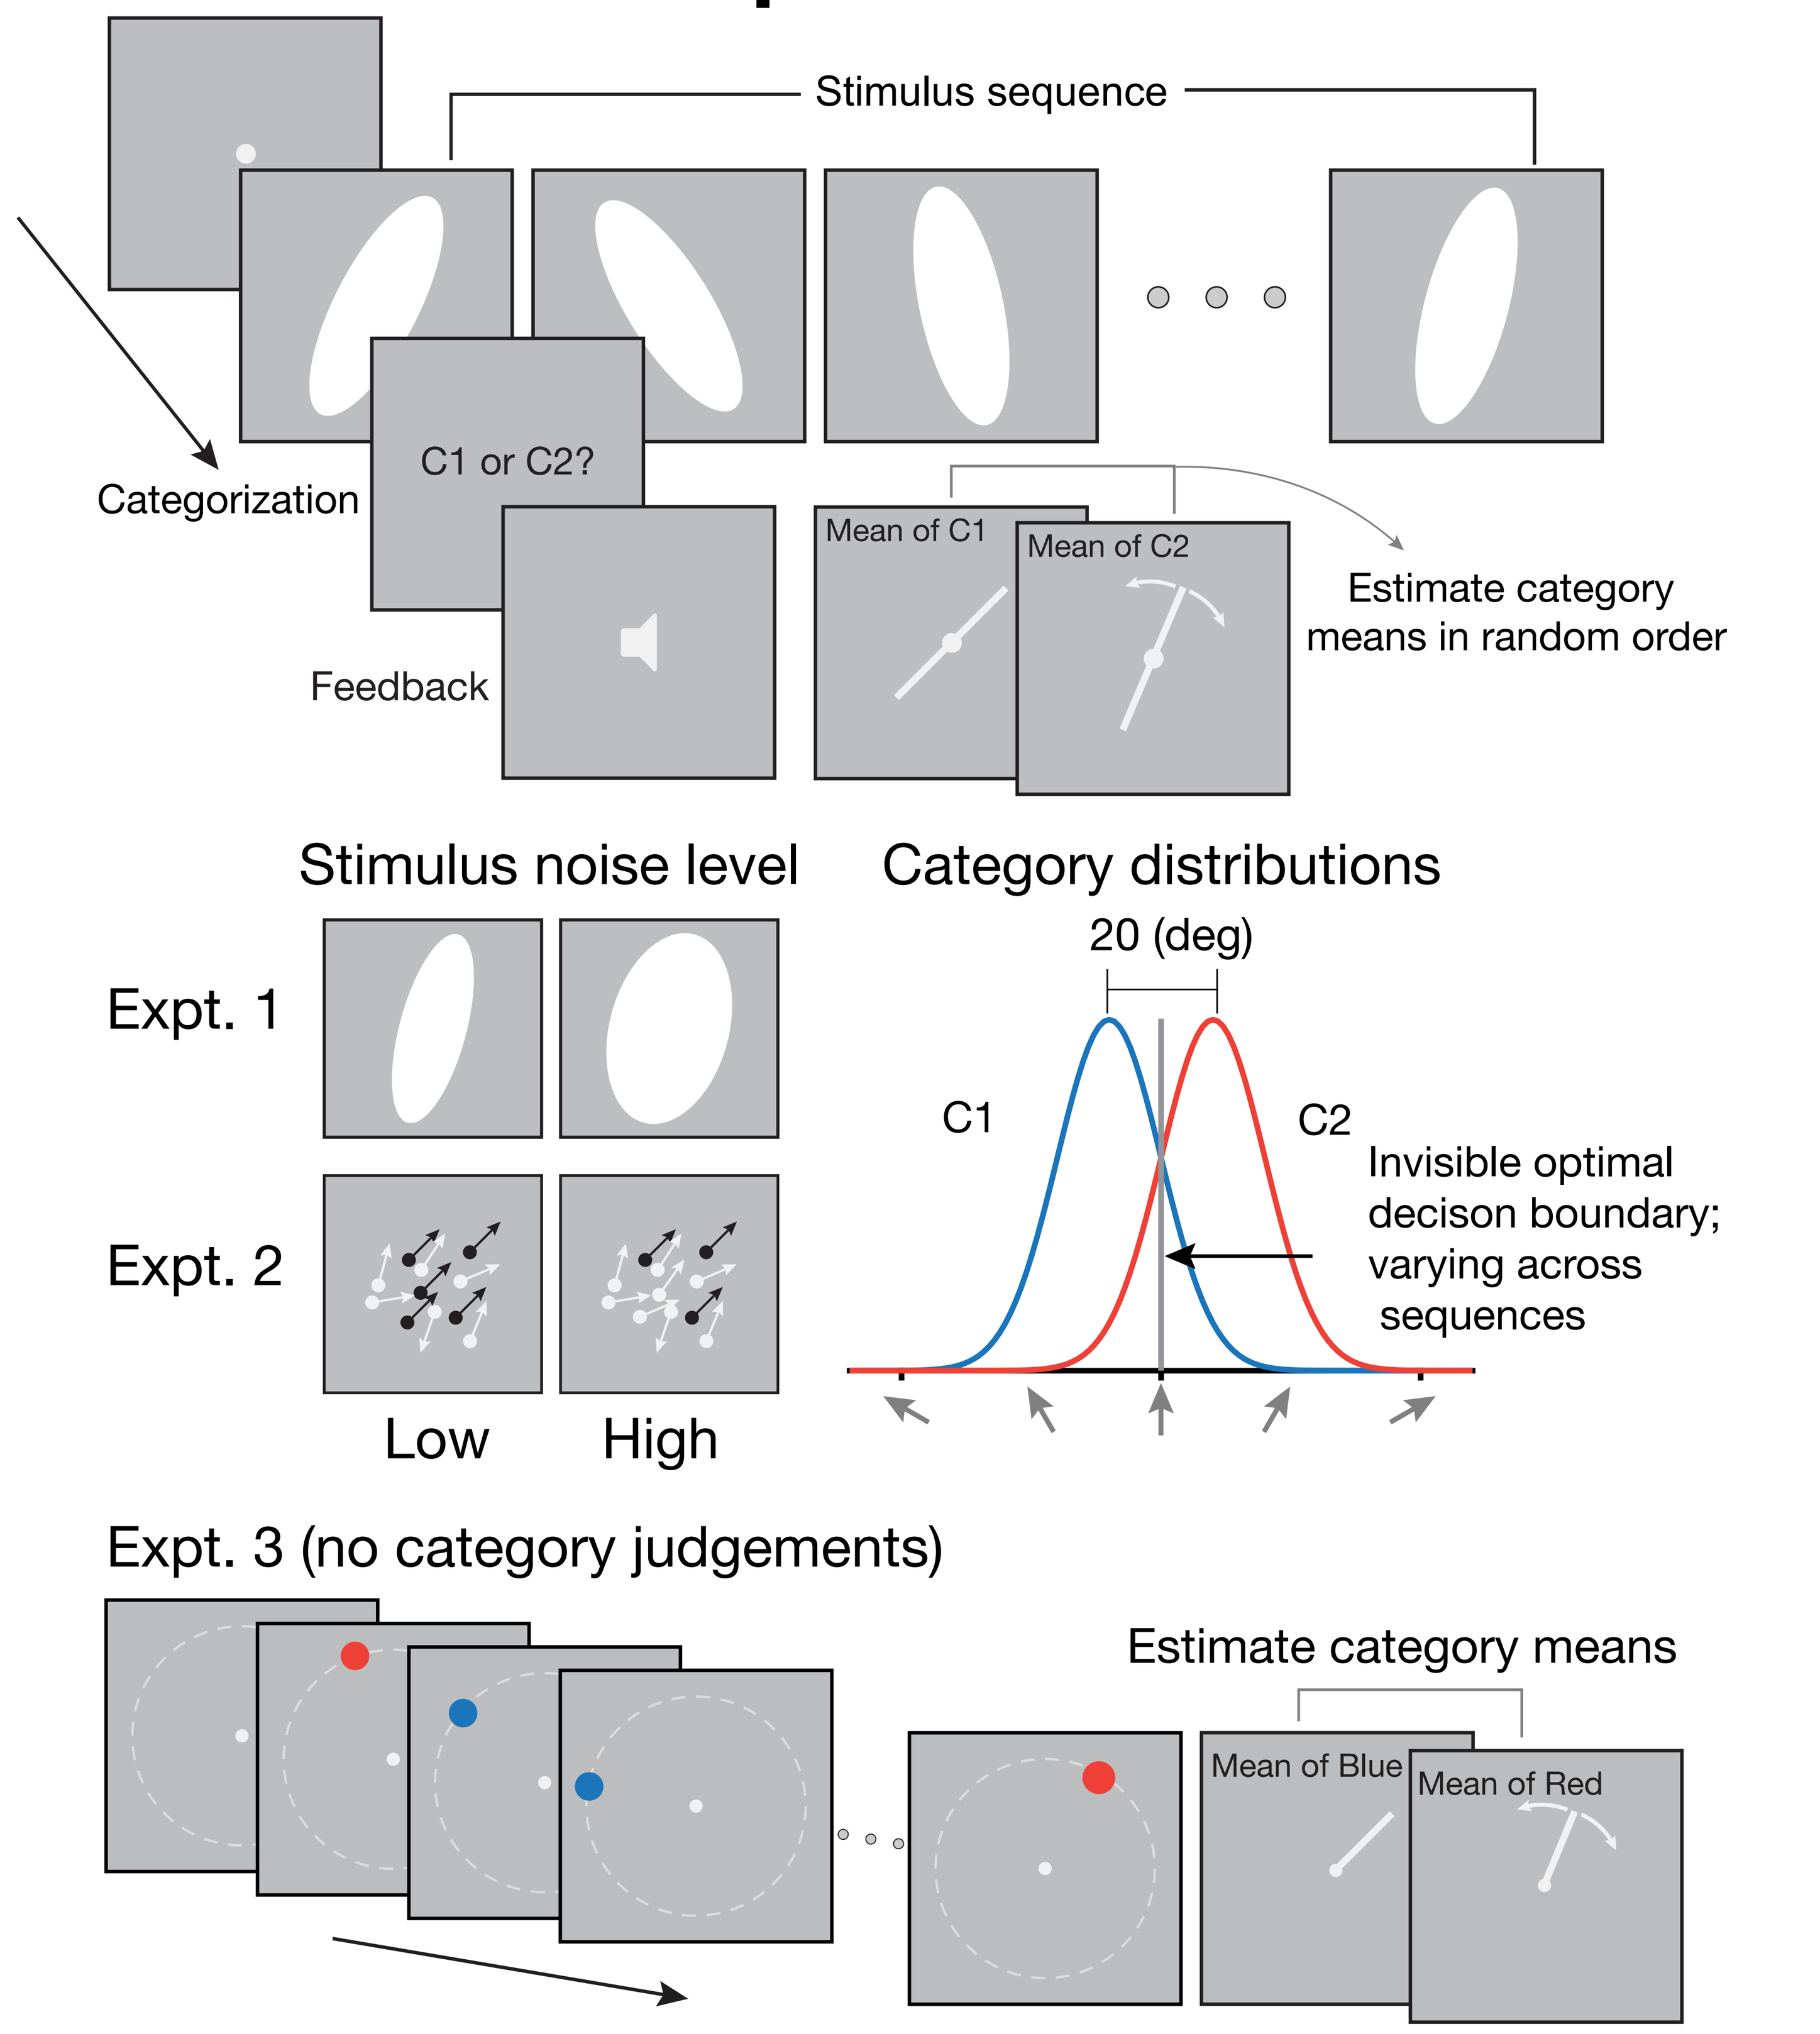
Stimulus sequence
Categorization
C1 or C2?
Feedback
Mean of C1
Mean of C2
Estimate category means in random order
Stimulus noise level
Category distributions
Expt. 1
Expt. 2
Low
High
20 (deg)
C1
C2
Invisible optimal decison boundary; varying across sequences
Expt. 3 (no category judgements)
Estimate category means
Mean of Blue
Mean of Red

Models/Hypotheses

Model 1
Self-consistent averaging
(Luu & Stocker, 2018)

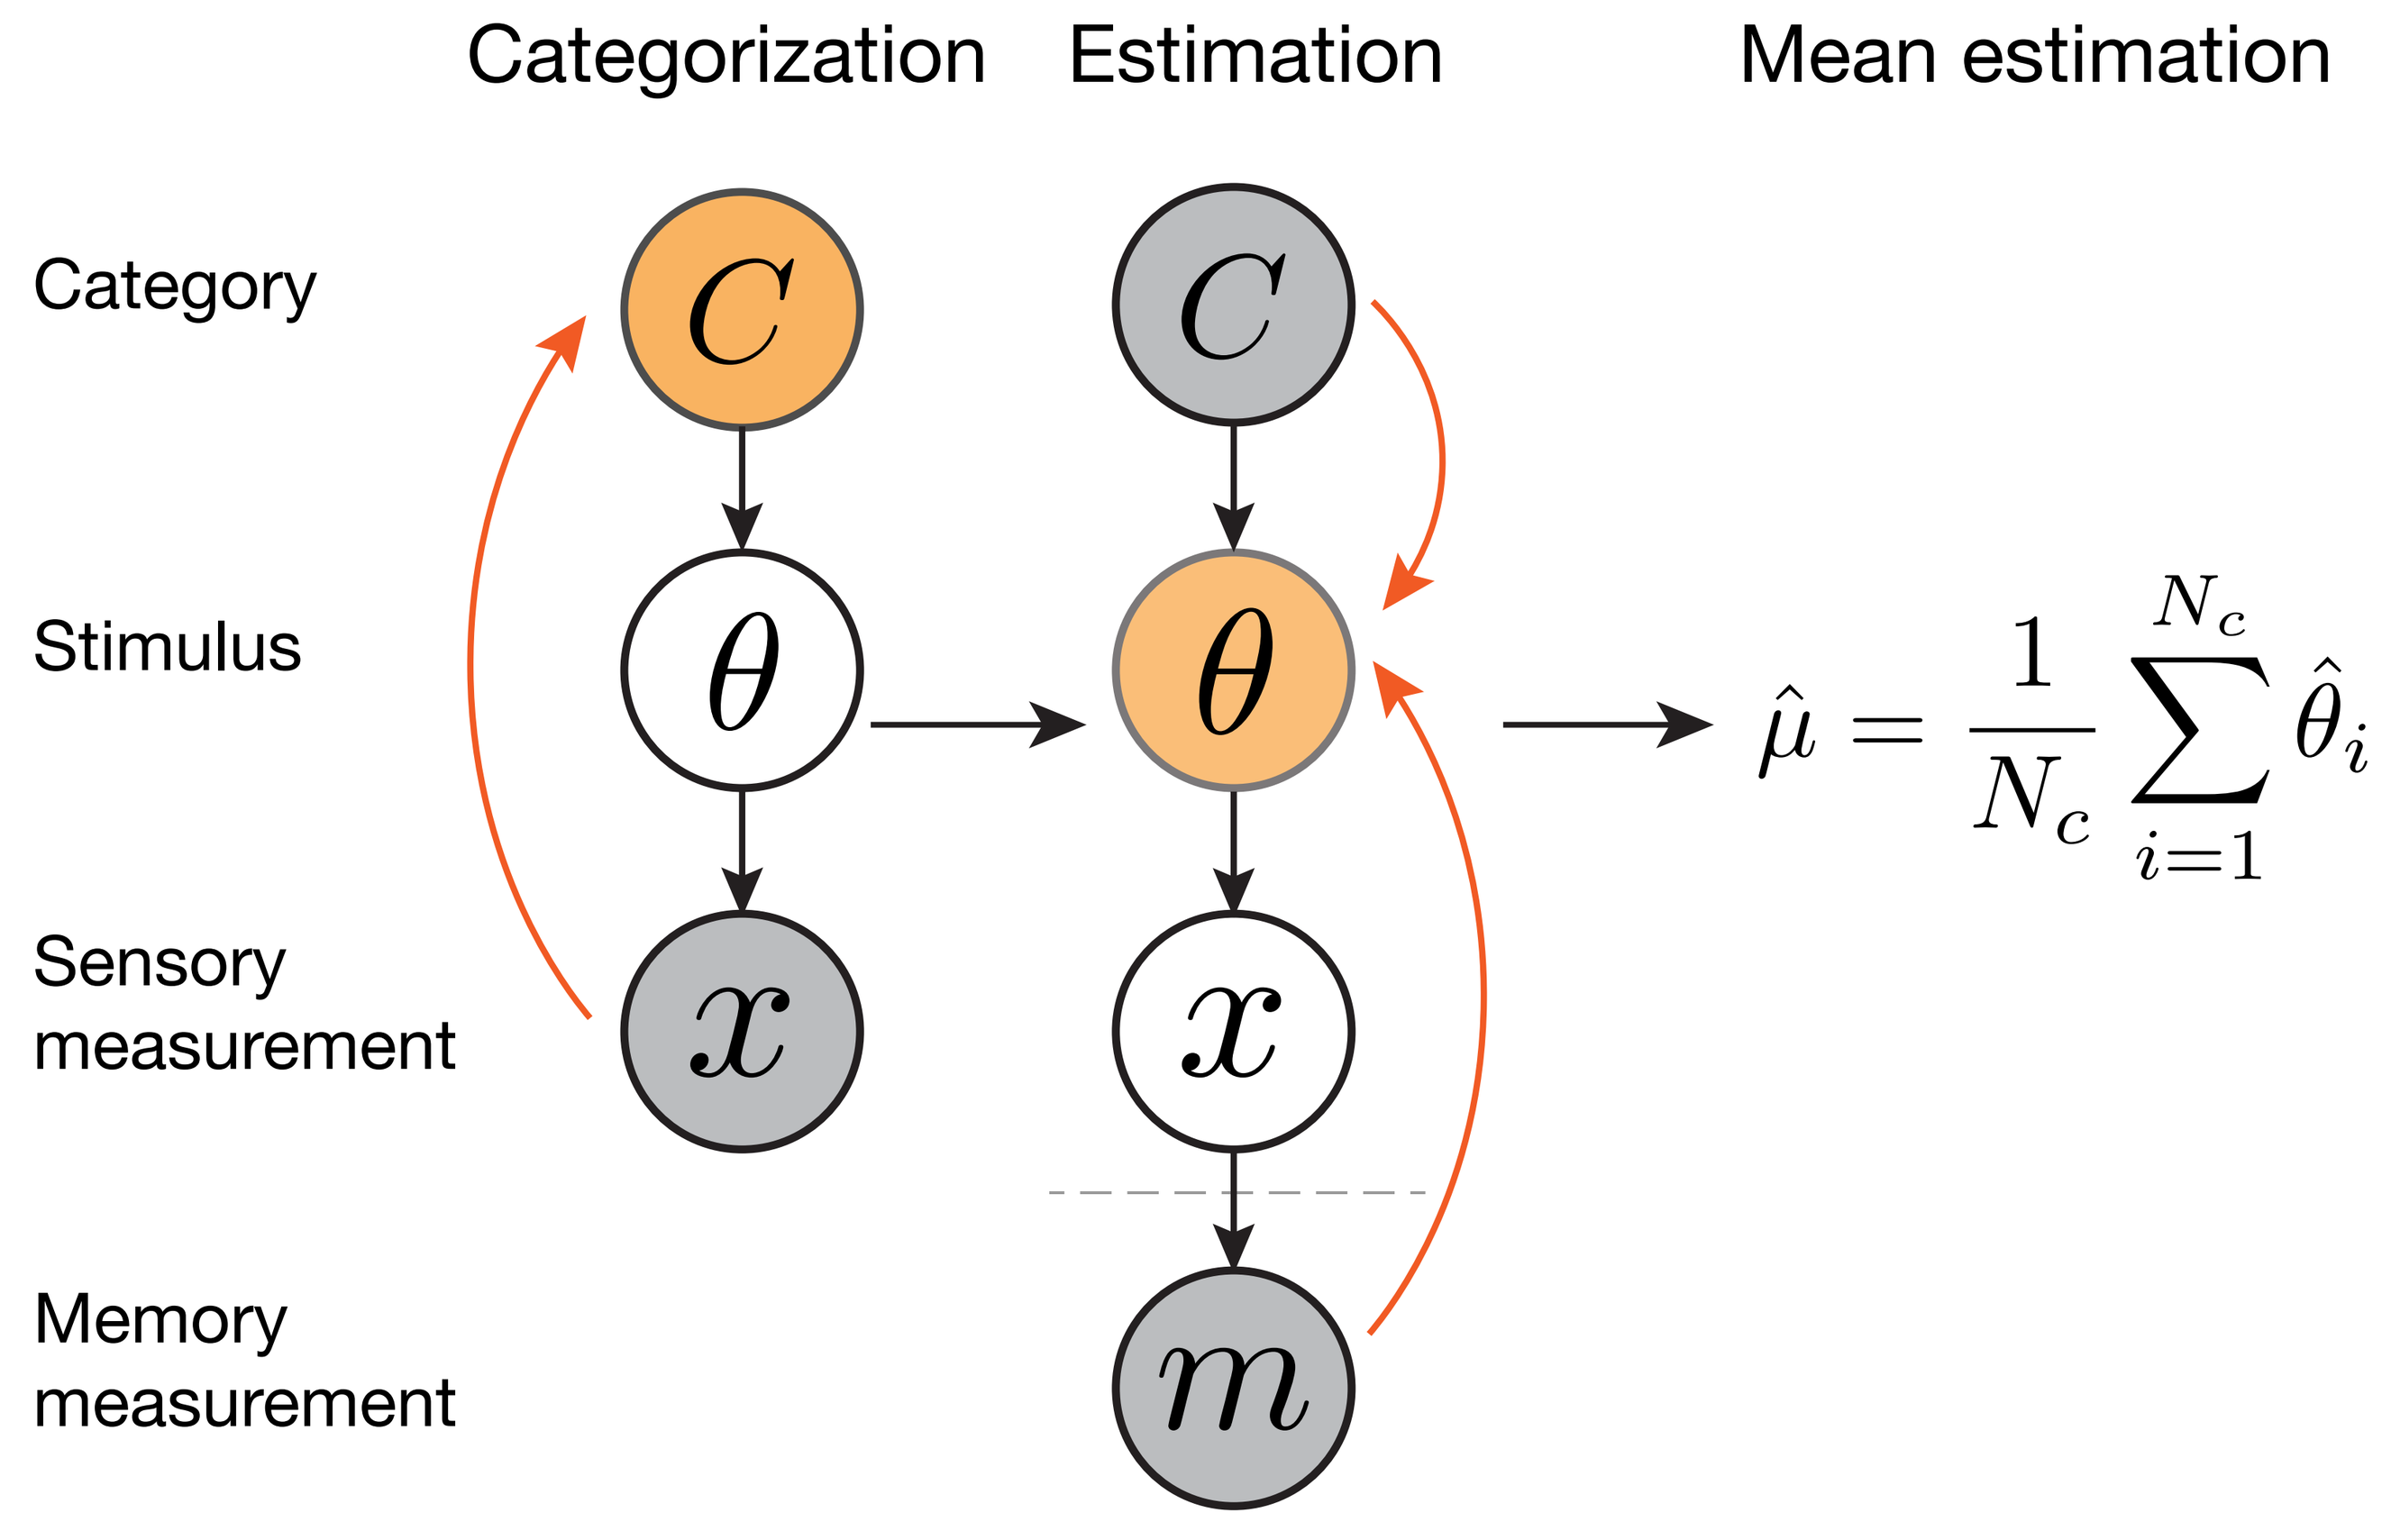
Categorization
Estimation
Mean estimation
Category
Stimulus
Sensory measurement
Memory measurement
$\hat{\mu} = \frac{1}{N_c}\sum_{i=1}^{N_c}\hat{\theta}_i$

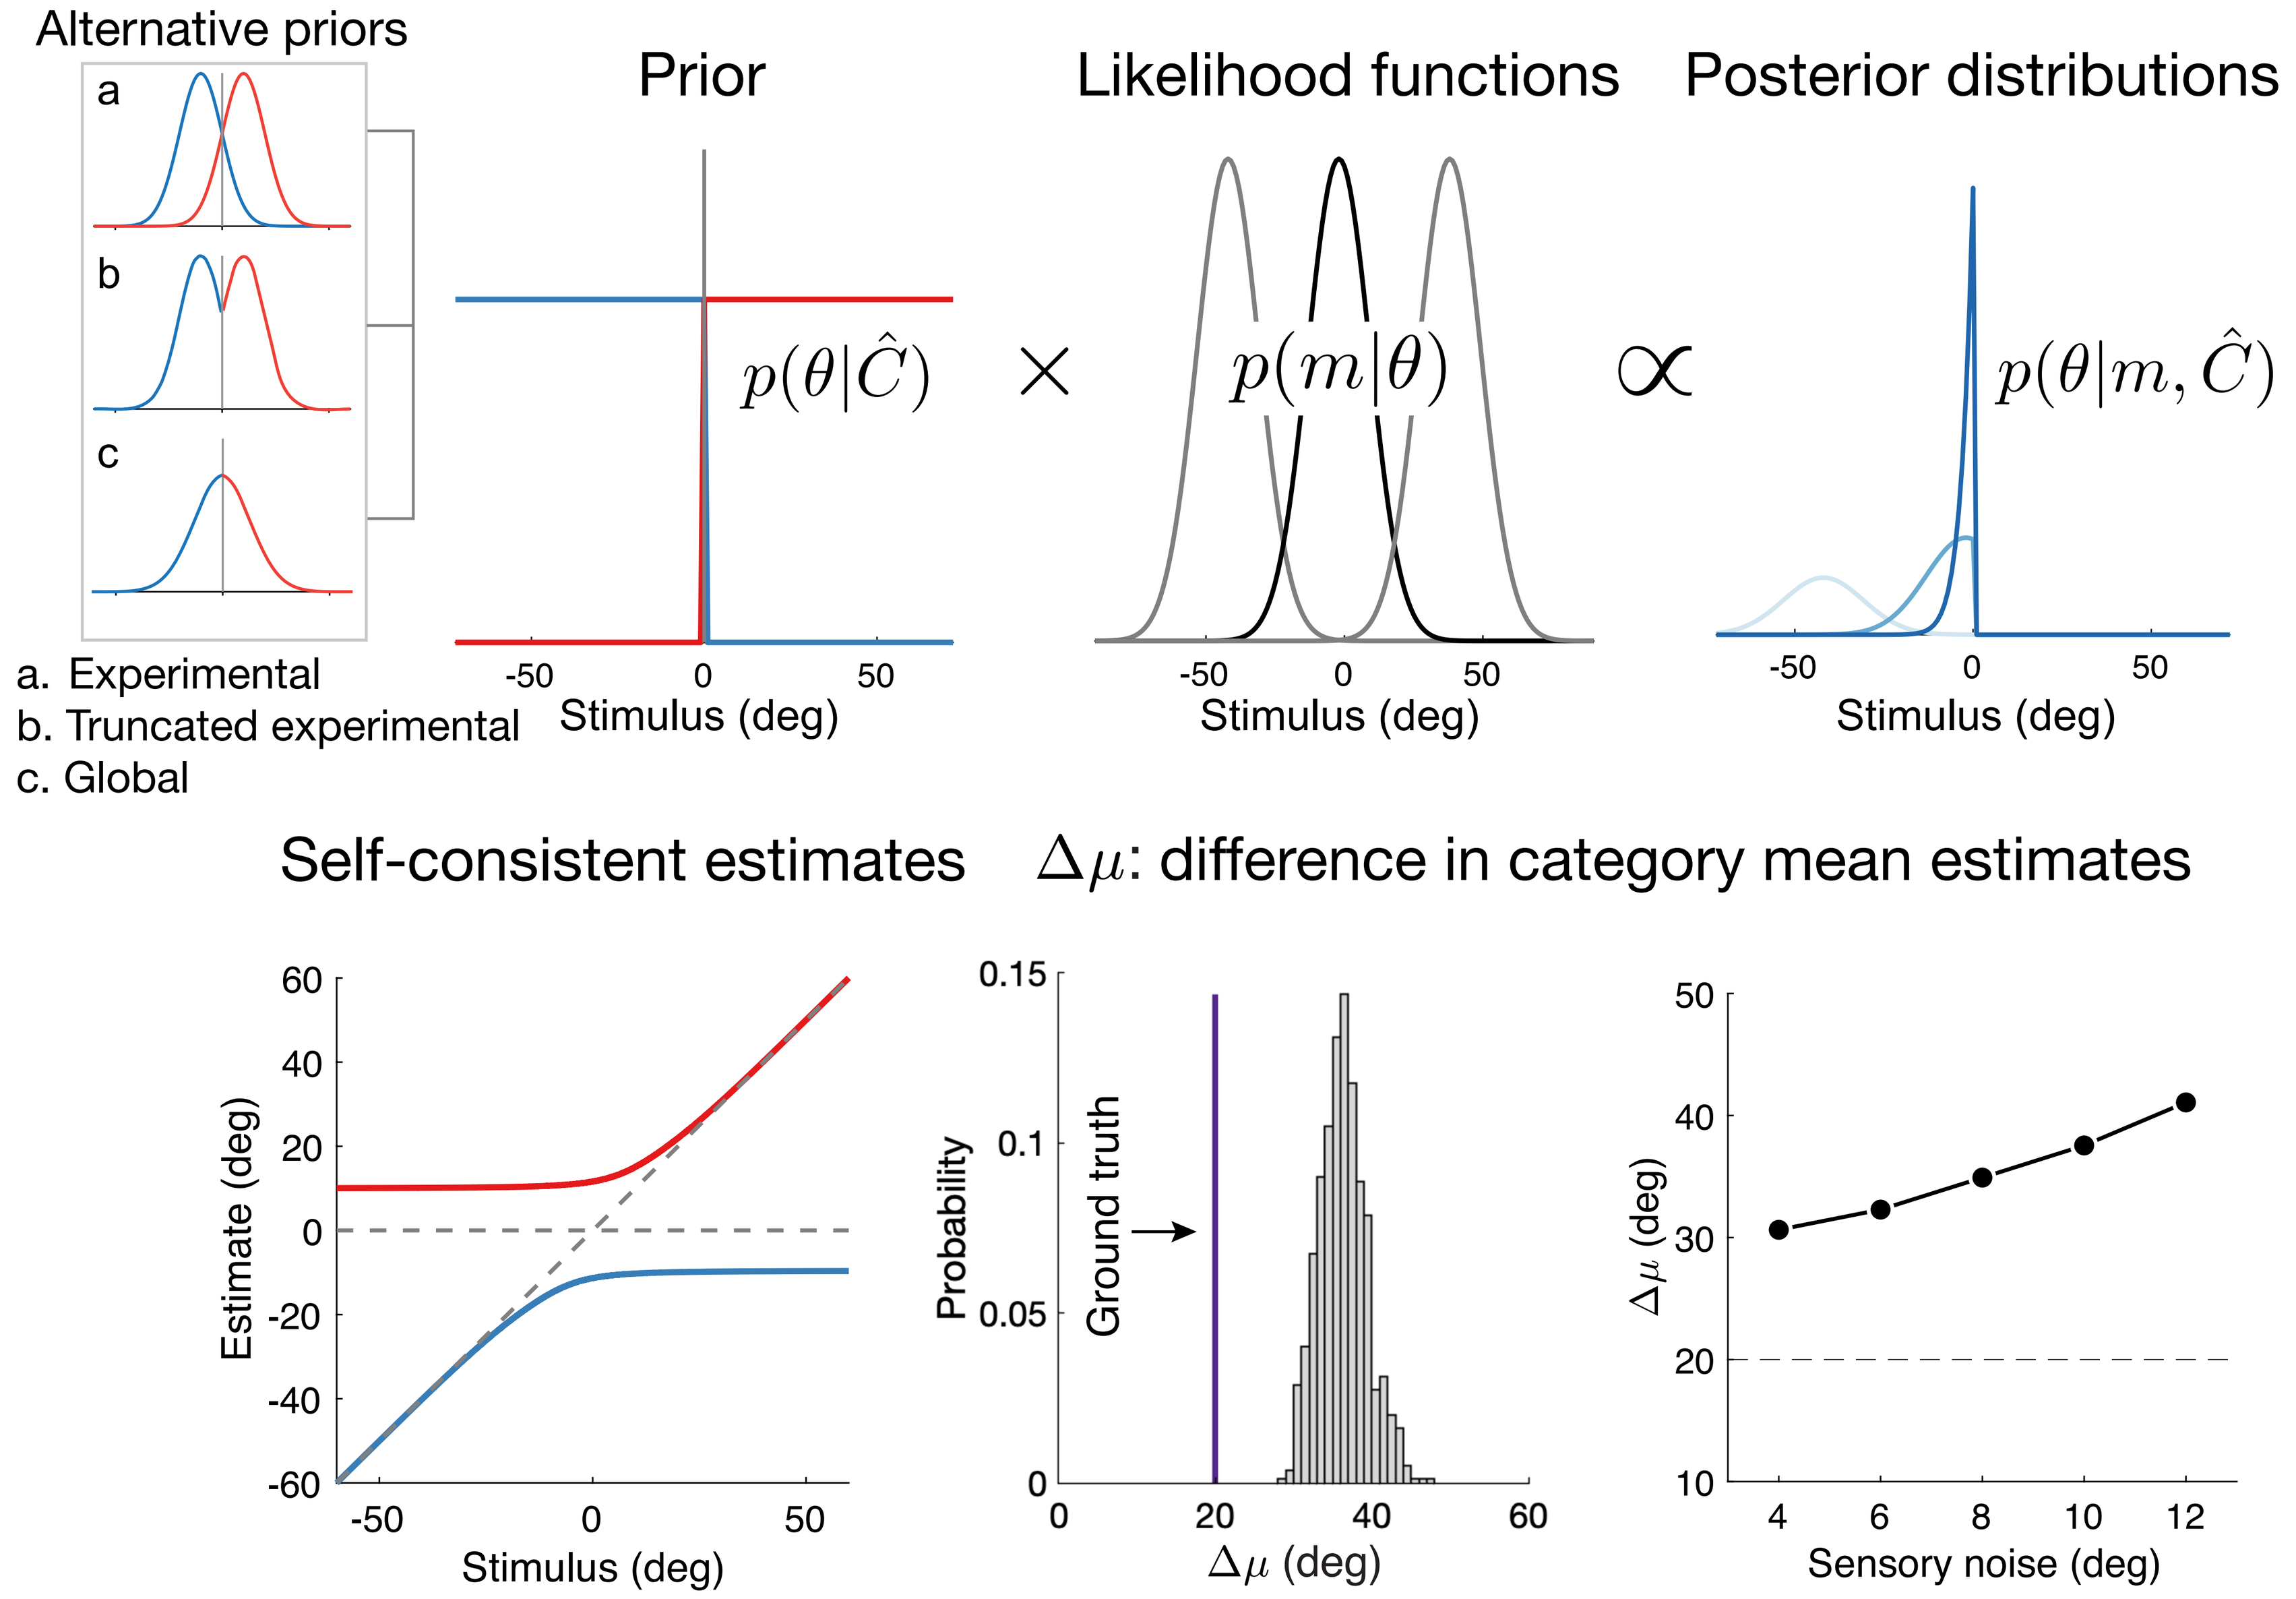
Alternative priors
a
b
c
a. Experimental
b. Truncated experimental
c. Global
Prior
Likelihood functions
Posterior distributions
$p(\theta|\hat{C})$
$p(m|\theta)$
$p(\theta|m,\hat{C})$
Stimulus (deg)
Self-consistent estimates
$\Delta\mu$: difference in category mean estimates
Estimate (deg)
Probability
Ground truth
$\Delta\mu$ (deg)
Sensory noise (deg)

Model 2
Hierarchical decoding
(Ding at el., 2017)

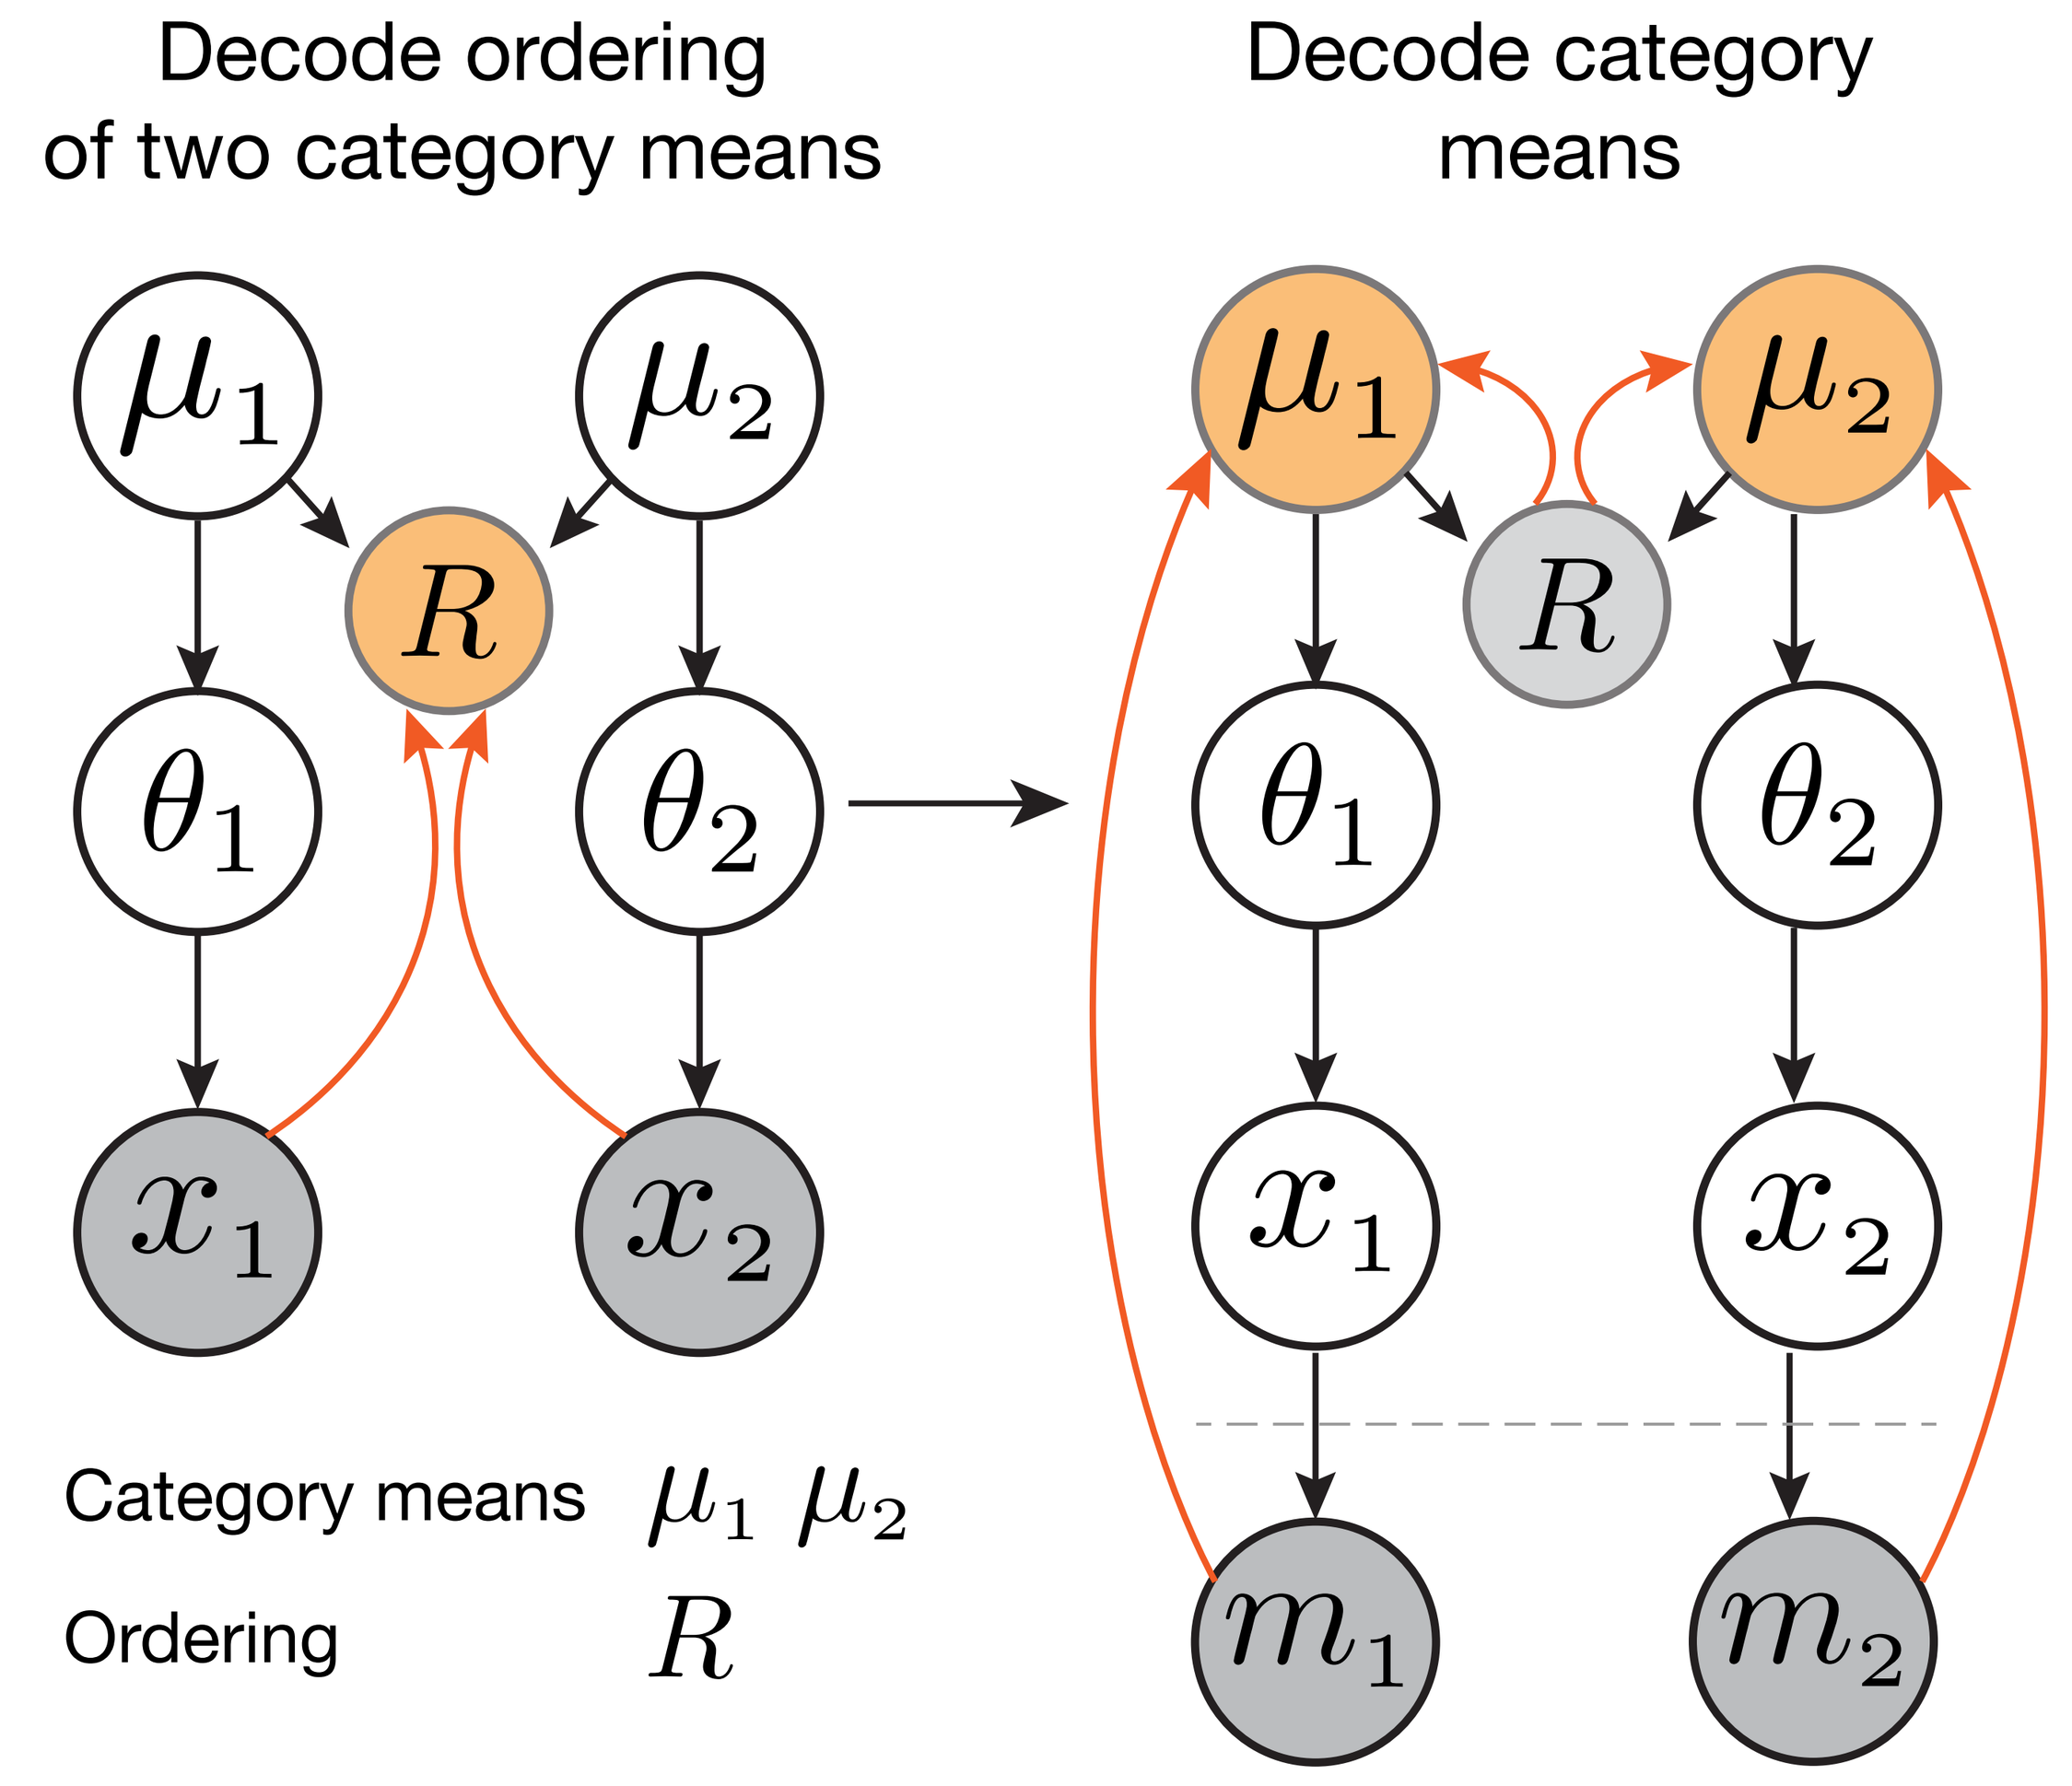
Decode ordering of two category means
Decode category means
Category means μ_1 μ_2
Ordering R

Hierarchical decoding repeats after each stimulus.

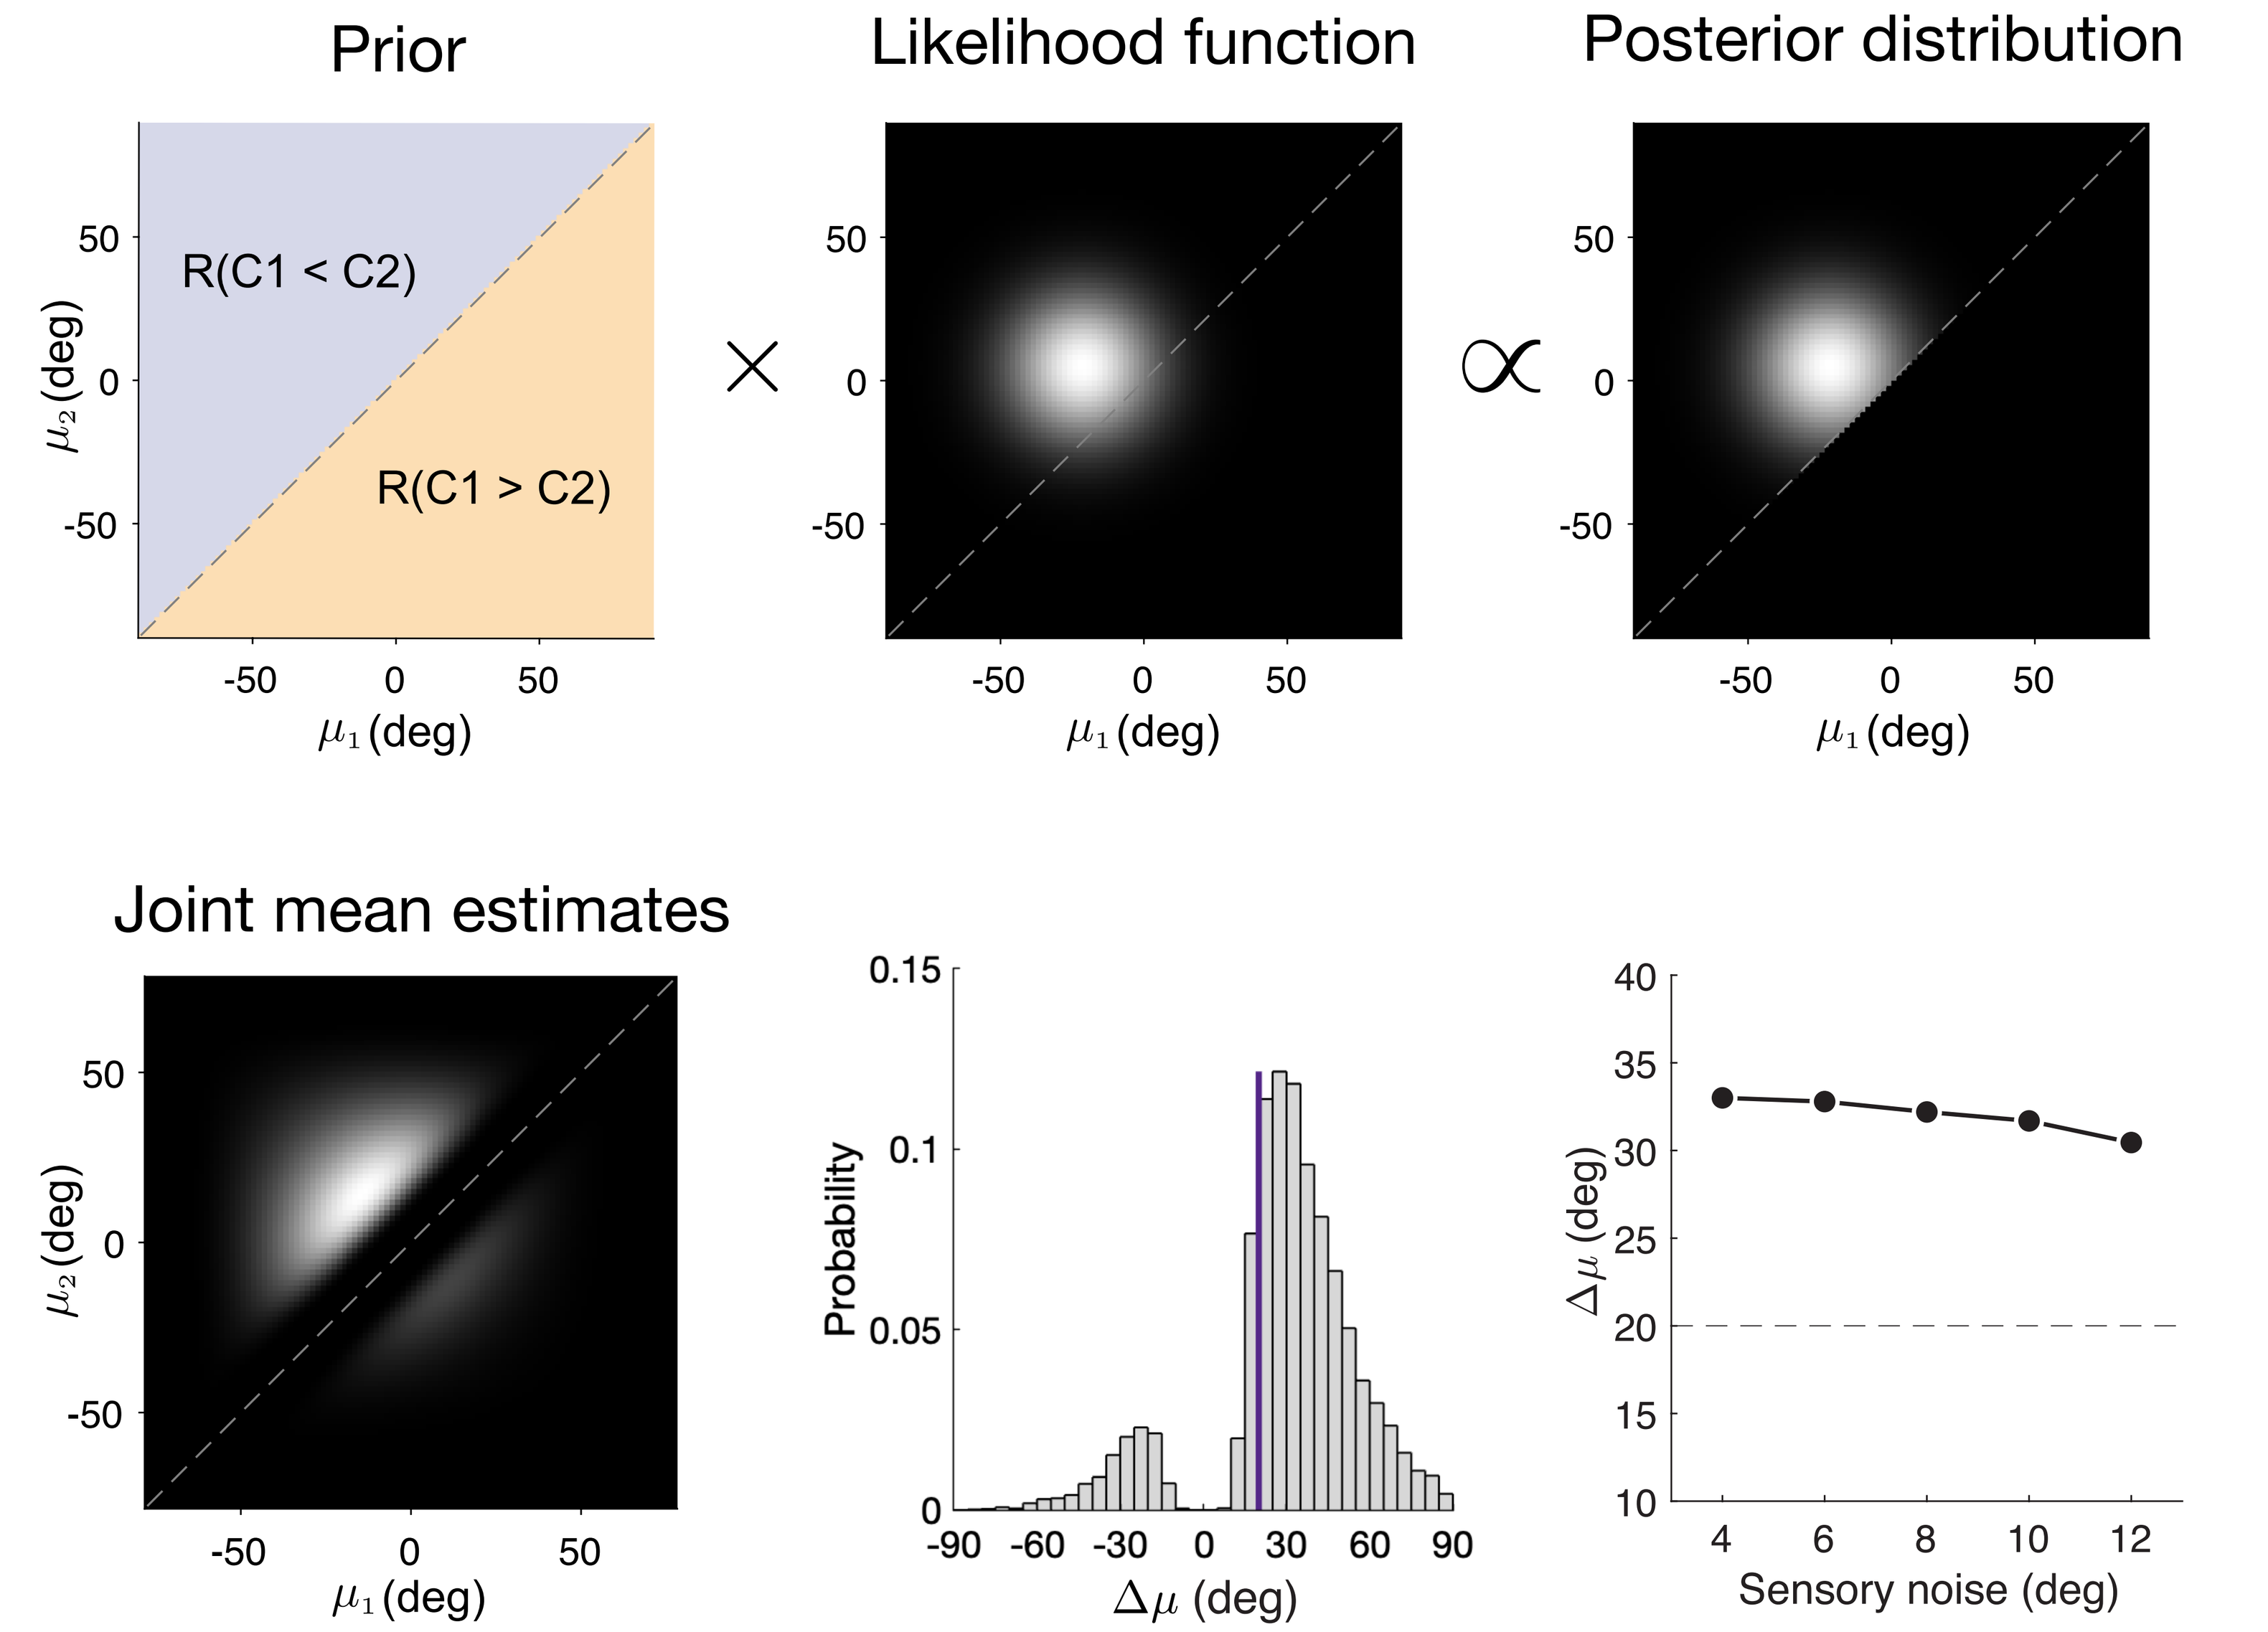
Prior
Likelihood function
Posterior distribution
R(C1 < C2)
R(C1 > C2)
μ_2 (deg)
μ_1 (deg)
Joint mean estimates
Probability
$\Delta\mu$ (deg)
Sensory noise (deg)

Results

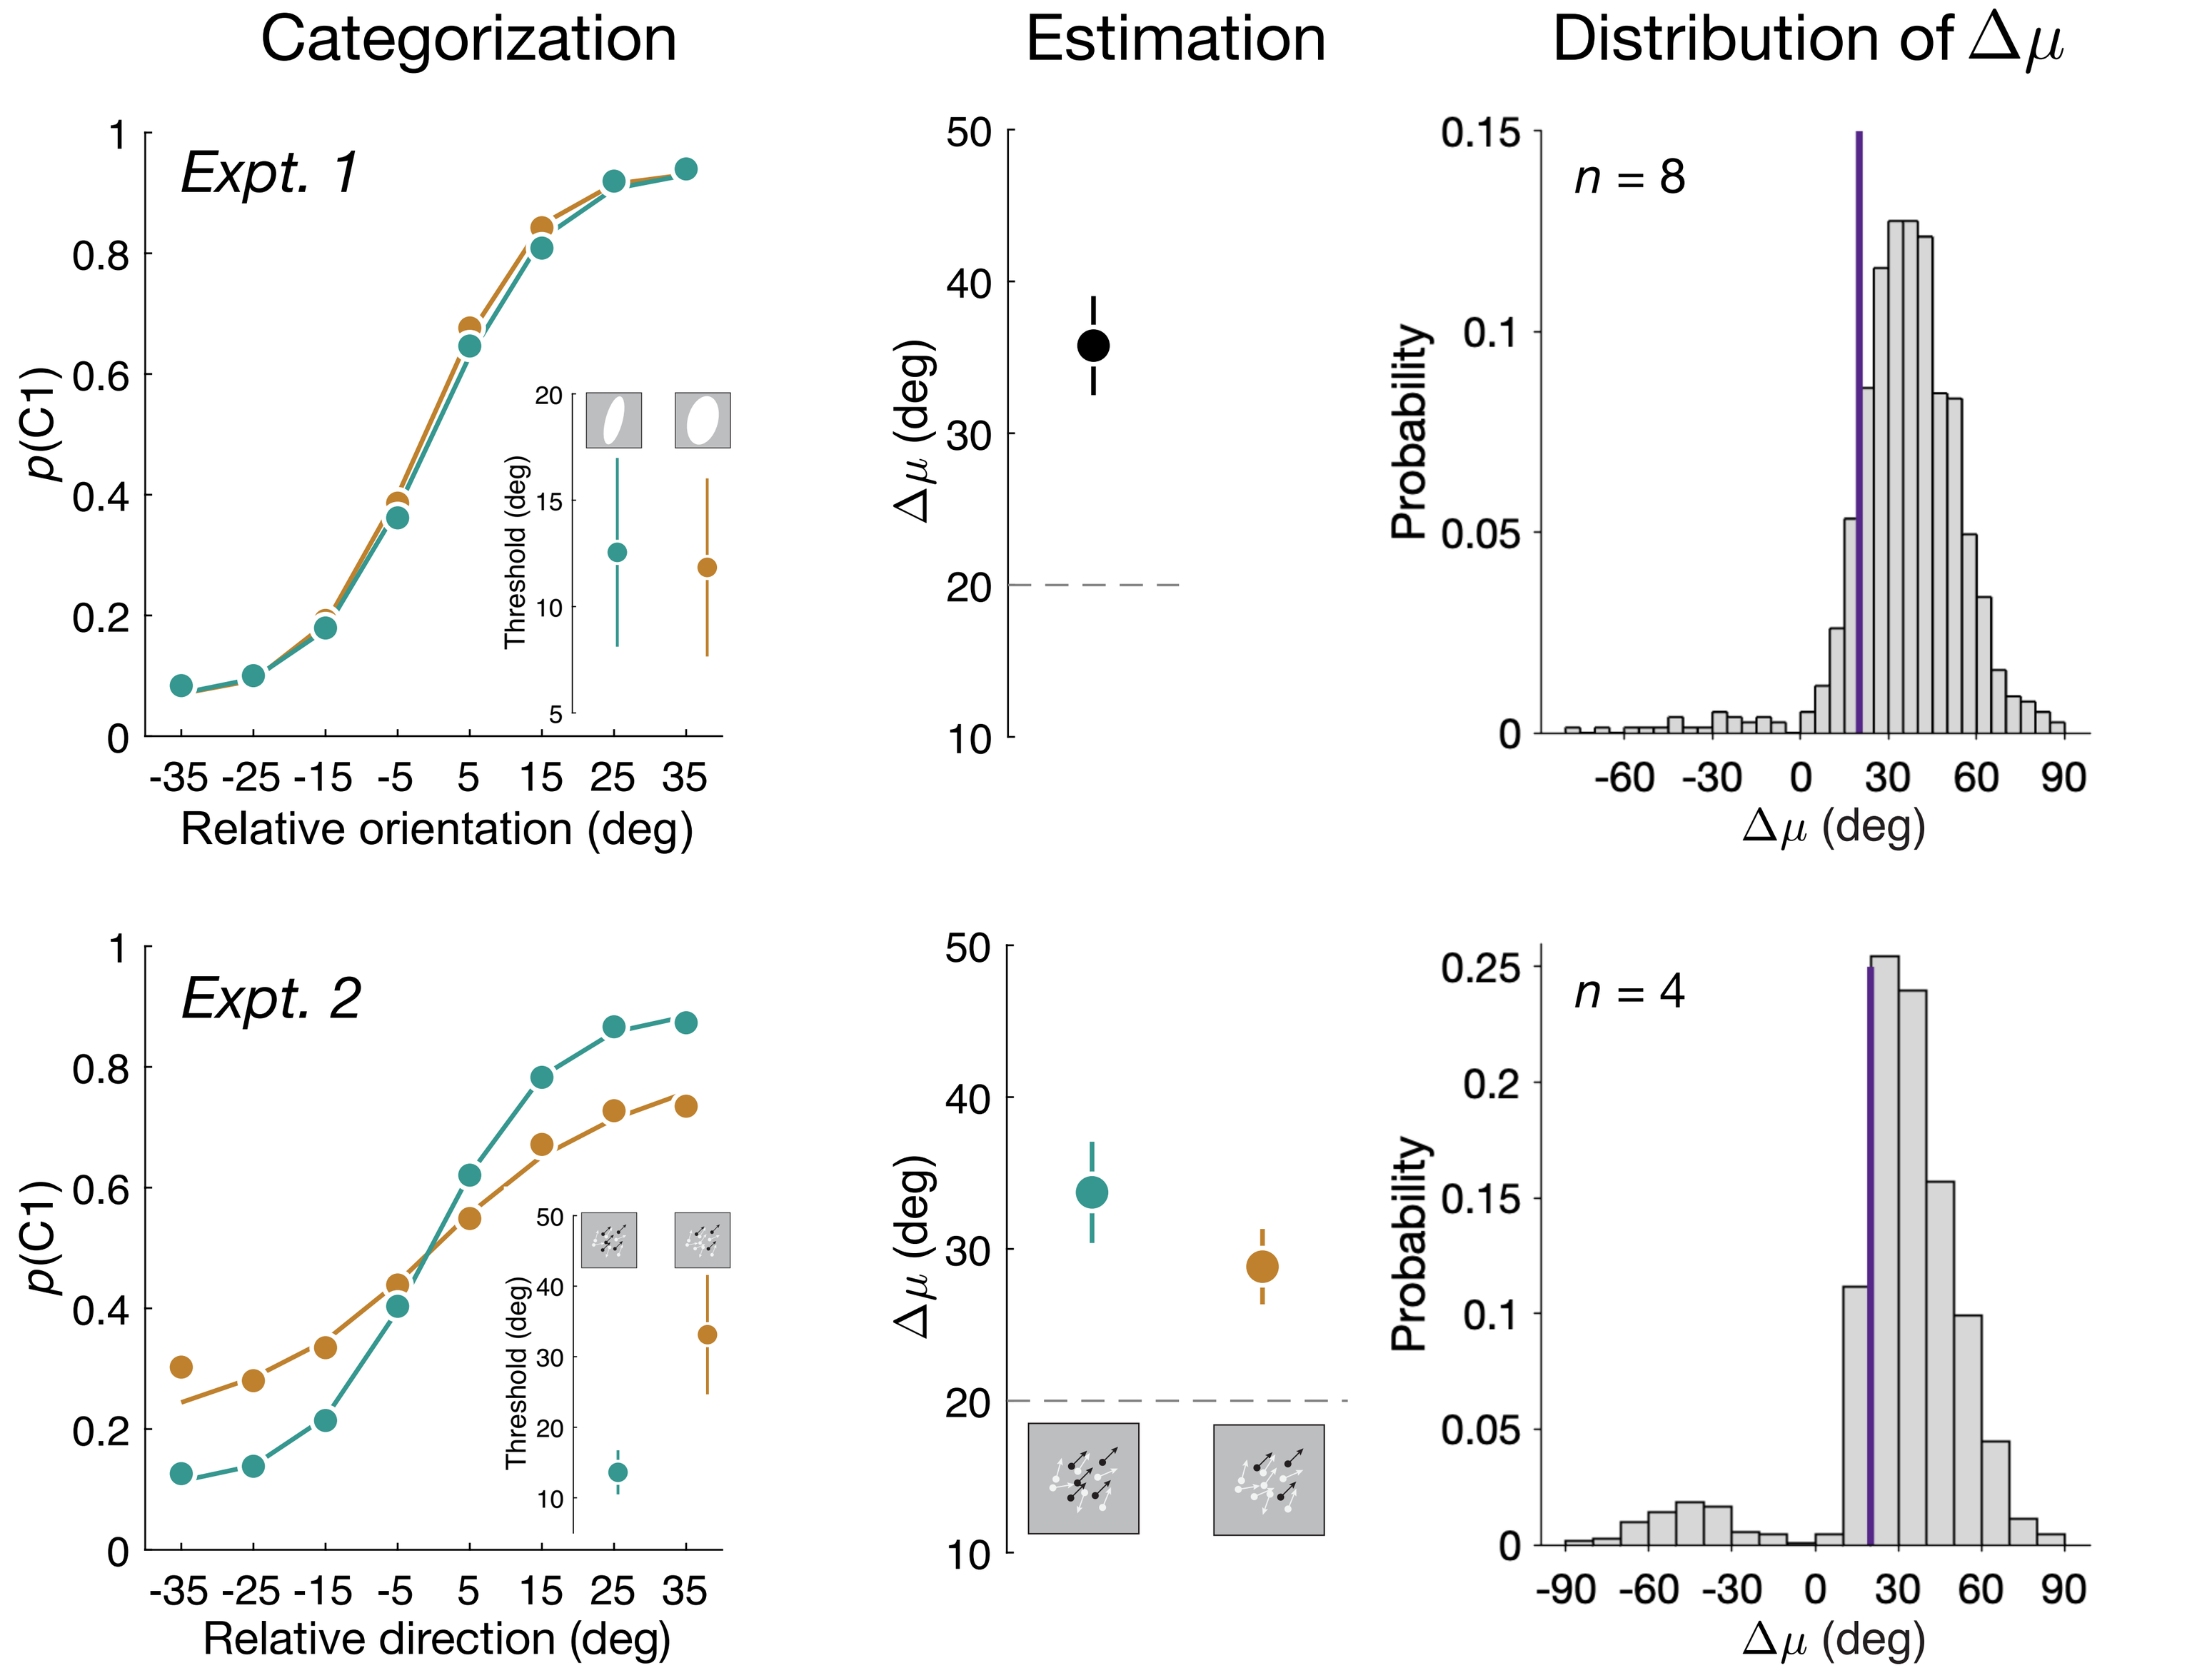
Categorization
Estimation
Distribution of $\Delta\mu$
Expt. 1
Expt. 2
p(C1)
Relative orientation (deg)
Relative direction (deg)
Threshold (deg)
$\Delta\mu$ (deg)
Probability
n = 8
n = 4

Highlights

- More sensory noise leads to less repulsion.
- Distribution of mean estimate differences is bimodal.
- Repulsion remains strong with no explicit category decisions.

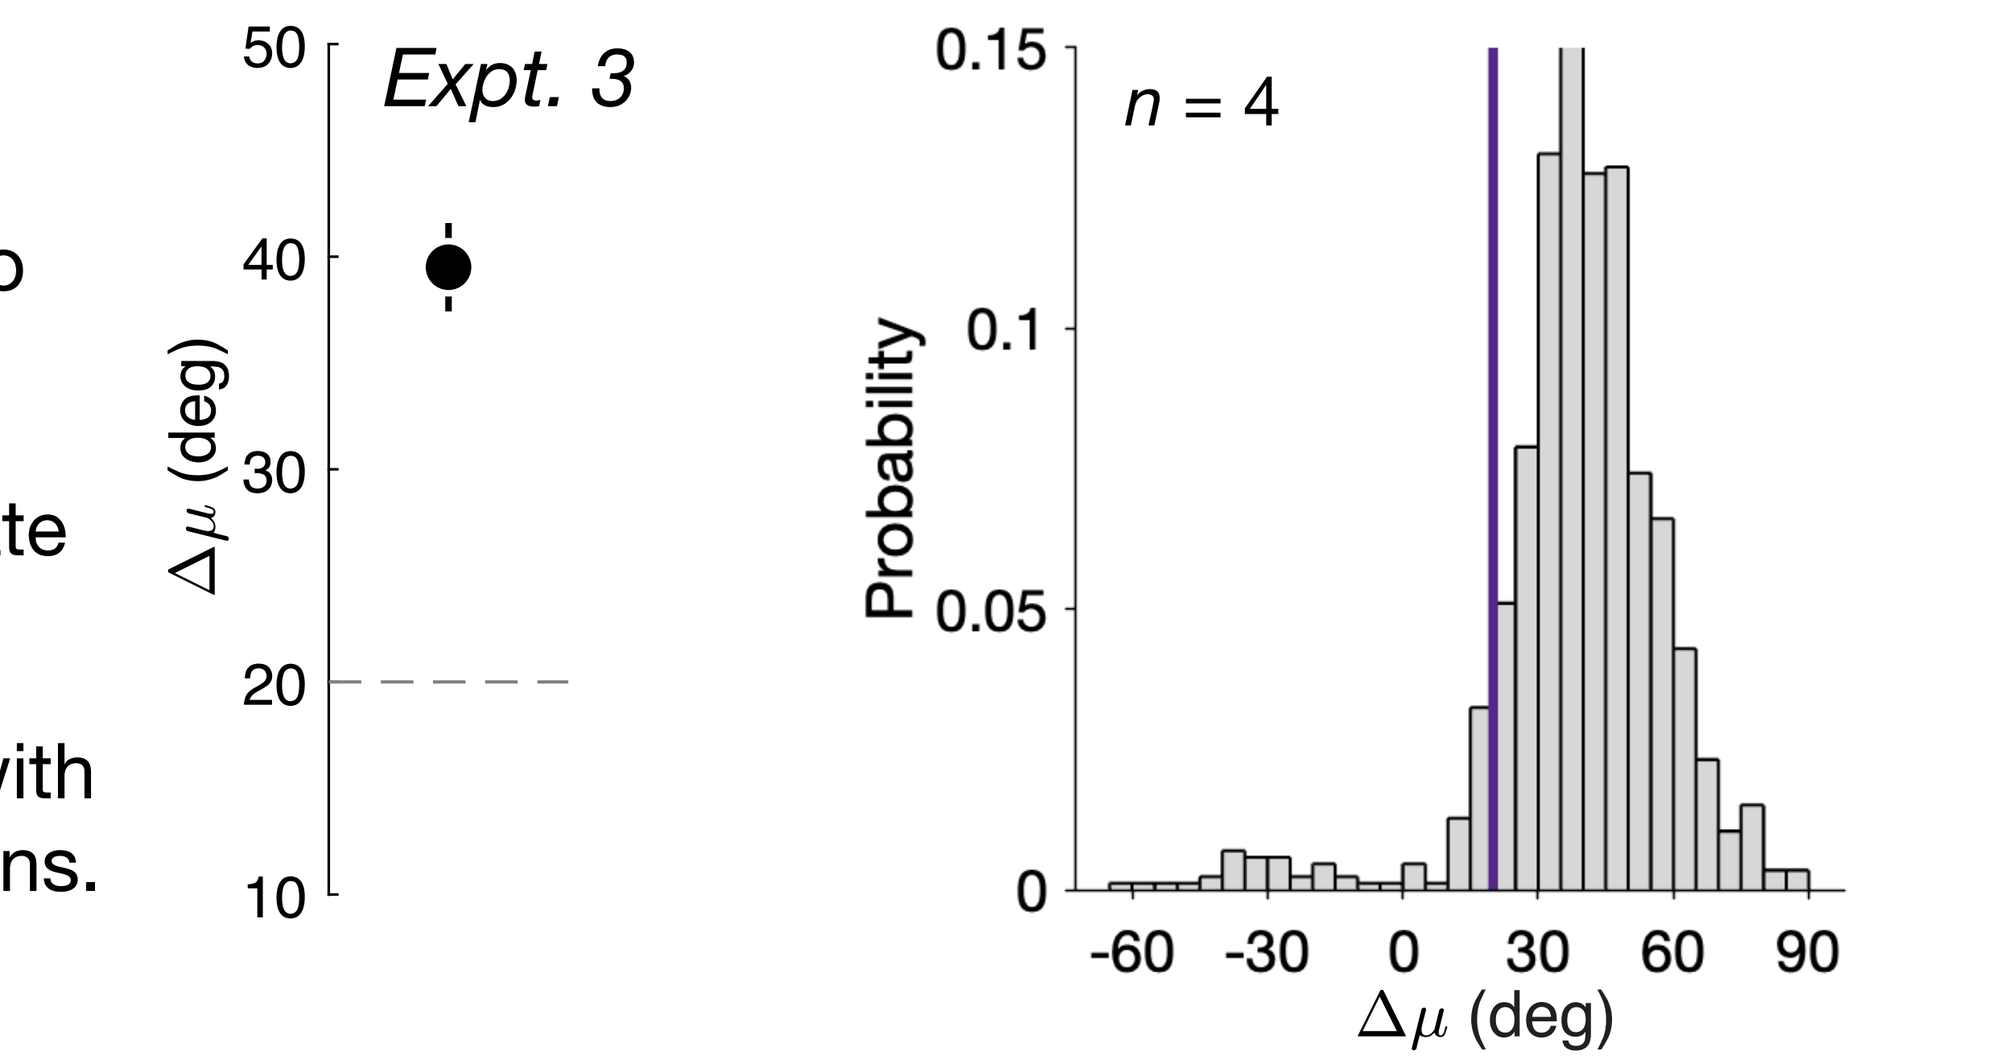
Expt. 3
$\Delta\mu$ (deg)
Probability
n = 4

Conclusion

Decoding proceeds hierarchically: observers first determine the ordering (e.g., cw/ccw) of the two categories, then use that order to constrain decoding of category means.

Luu, L., & Stocker, A. A. (2018). Post-decision biases reveal a self-consistency principle in perceptual inference. *Elife, 7*, e33334.

Ding, S., Cueva, C. J., Tsodyks, M., & Qian, N. (2017). Visual perception as retrospective Bayesian decoding from high-to low-level features, *PNAS, 114*(43), E9115-E9124.

Funding: NIH EY08266